Development of a Psoriasis PhysioPD™ Platform to Evaluate a Novel Therapy and Identify Uncertainties Critical to Efficacy and Competitive Differentiation

Katherine Kudrycki[1]*, Michael Weis[1], Rebecca Baillie[1], Michael Reed[1], Christiane Simon-Hinsberg[2], Hannelore Stockhausen[2], Silvia von Pistor[2], Anu Jauhan[2], Anna Dari[2], Sebastian Wachten[2], Marcel Froehlich[2], Wolfgang Prange[2], Roberta Bursi[2], Philipp Wabnitz[2]

*[1] Rosa and Co., San Carlos, CA, USA; [2] Grünenthal GmbH, Aachen, Germany *kkudrycki@rosaandco.com*

Introduction

- Psoriasis is a chronic, debilitating autoimmune skin disease affecting approximately 2% of the population with itching, thickened, red, scaly skin; 20-30% of psoriatic patients may develop psoriatic arthritis
- There are numerous anti-psoriatic treatments; however, there is widespread under-treatment due to lack/loss of response, safety concerns, and development of treatment tolerance
- Novel, orally delivered drugs with few side effects may help to overcome some of these obstacles
- A small molecule that fulfills these therapeutic criteria has been identified, but the specific enzyme isoform responsible for the mechanisms of action (MOA) remains uncertain
- Combining molecular biology, gene expression, and clinical data with computational mechanistic modeling (QSP) facilitated evaluation of the role of specific isoforms in the disease and treatment response

Objectives

- Identify mechanistic differences between the effects of **pan-enzyme inhibition** and **isoform-specific inhibition** on the disease response
- Provide insight into the **therapeutic potential of isoform-specific inhibitors**
- Evaluate the potential efficacy of a select **isoform-specific inhibitor,** a candidate for **the novel treatment**
- Reduce risk for future stages of research and development by gaining new insights into the pathophysiology of psoriasis

Methods

The Psoriasis PhysioPD™ Research Platform is a mechanistic, quantitative model representing key biological mechanisms involved in the pathophysiology of psoriasis and response to treatments.

- The Psoriasis PhysioPD Platform is a mathematical representation of a single moderate-to-severe psoriatic plaque
- The Platform represents the dynamics of keratinocytes, immune cells, key cytokines and chemokines, as well as the pharmacokinetics and pharmacodynamics of various therapies (Figure 2)
- Data and scientific knowledge from numerous sources are integrated into a single contextual framework
- Rosa's Model Qualification Method was used to ensure the model was fit-for-purpose (Figure 1)
- Key drivers of treatment efficacy were identified by **sensitivity analysis** (SA)
- **Virtual Patients** (VPs) were created to explore the impact of target-specific uncertainties
- **Target-specific uncertainties** were identified from a literature survey of enzyme subtype expression/function

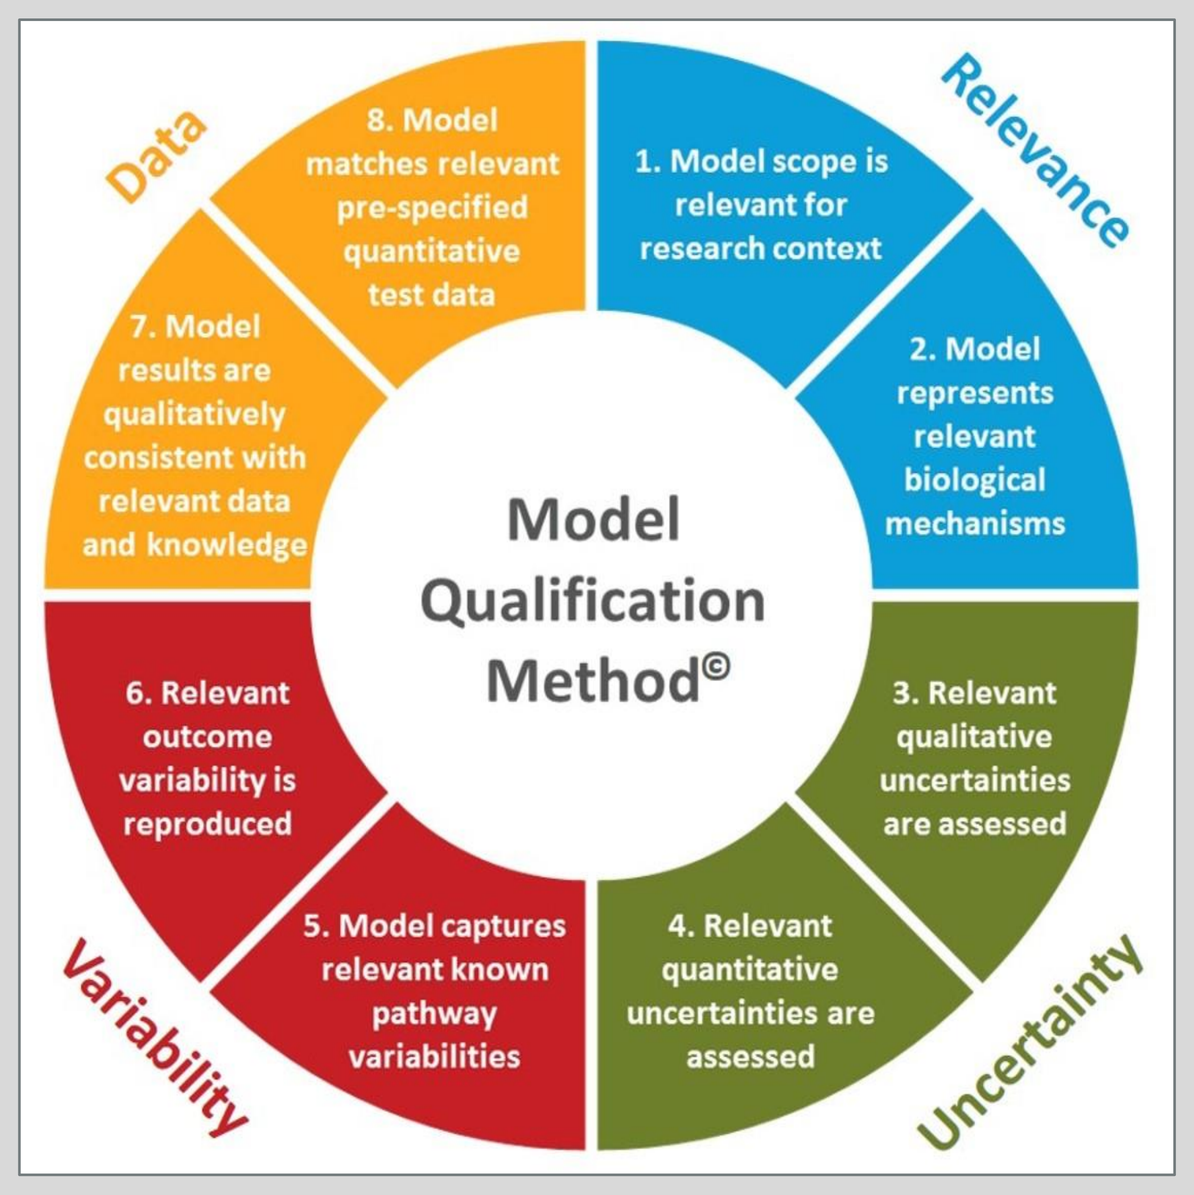
Data
Relevance
Uncertainty
Variability
Model Qualification Method©
1. Model scope is relevant for research context
2. Model represents relevant biological mechanisms
3. Relevant qualitative uncertainties are assessed
4. Relevant quantitative uncertainties are assessed
5. Model captures relevant known pathway variabilities
6. Relevant outcome variability is reproduced
7. Model results are qualitatively consistent with relevant data and knowledge
8. Model matches relevant pre-specified quantitative test data

Figure 1. Diagram of Rosa's Model Qualification Method[1] (MQM)

References

1. Friedrich, CM. (2016) CPT: Pharmacometrics & Systems Pharmacology 5, 43-53
2. Schmidt H, Jirstrand M. (2006) Bioinformatics 22, 514-515 (SimBiology)
3. Micali et al. (2016) Skin Research & Technology, 22, 341-348
4. Reddy et al. (2010) J Dermatol 37, 413-425
5. Rios-Navarro et al. (2015) Eur J Physiol 765, 355-365
6. Thaci et al. (2014) j Am Academ Dermatol 73, 400-409

Results

The Psoriasis PhysioPD Research Platform represents the biological components involved the pathogenesis of psoriasis, mechanisms of anti-psoriatic therapies, and clinical outcomes.

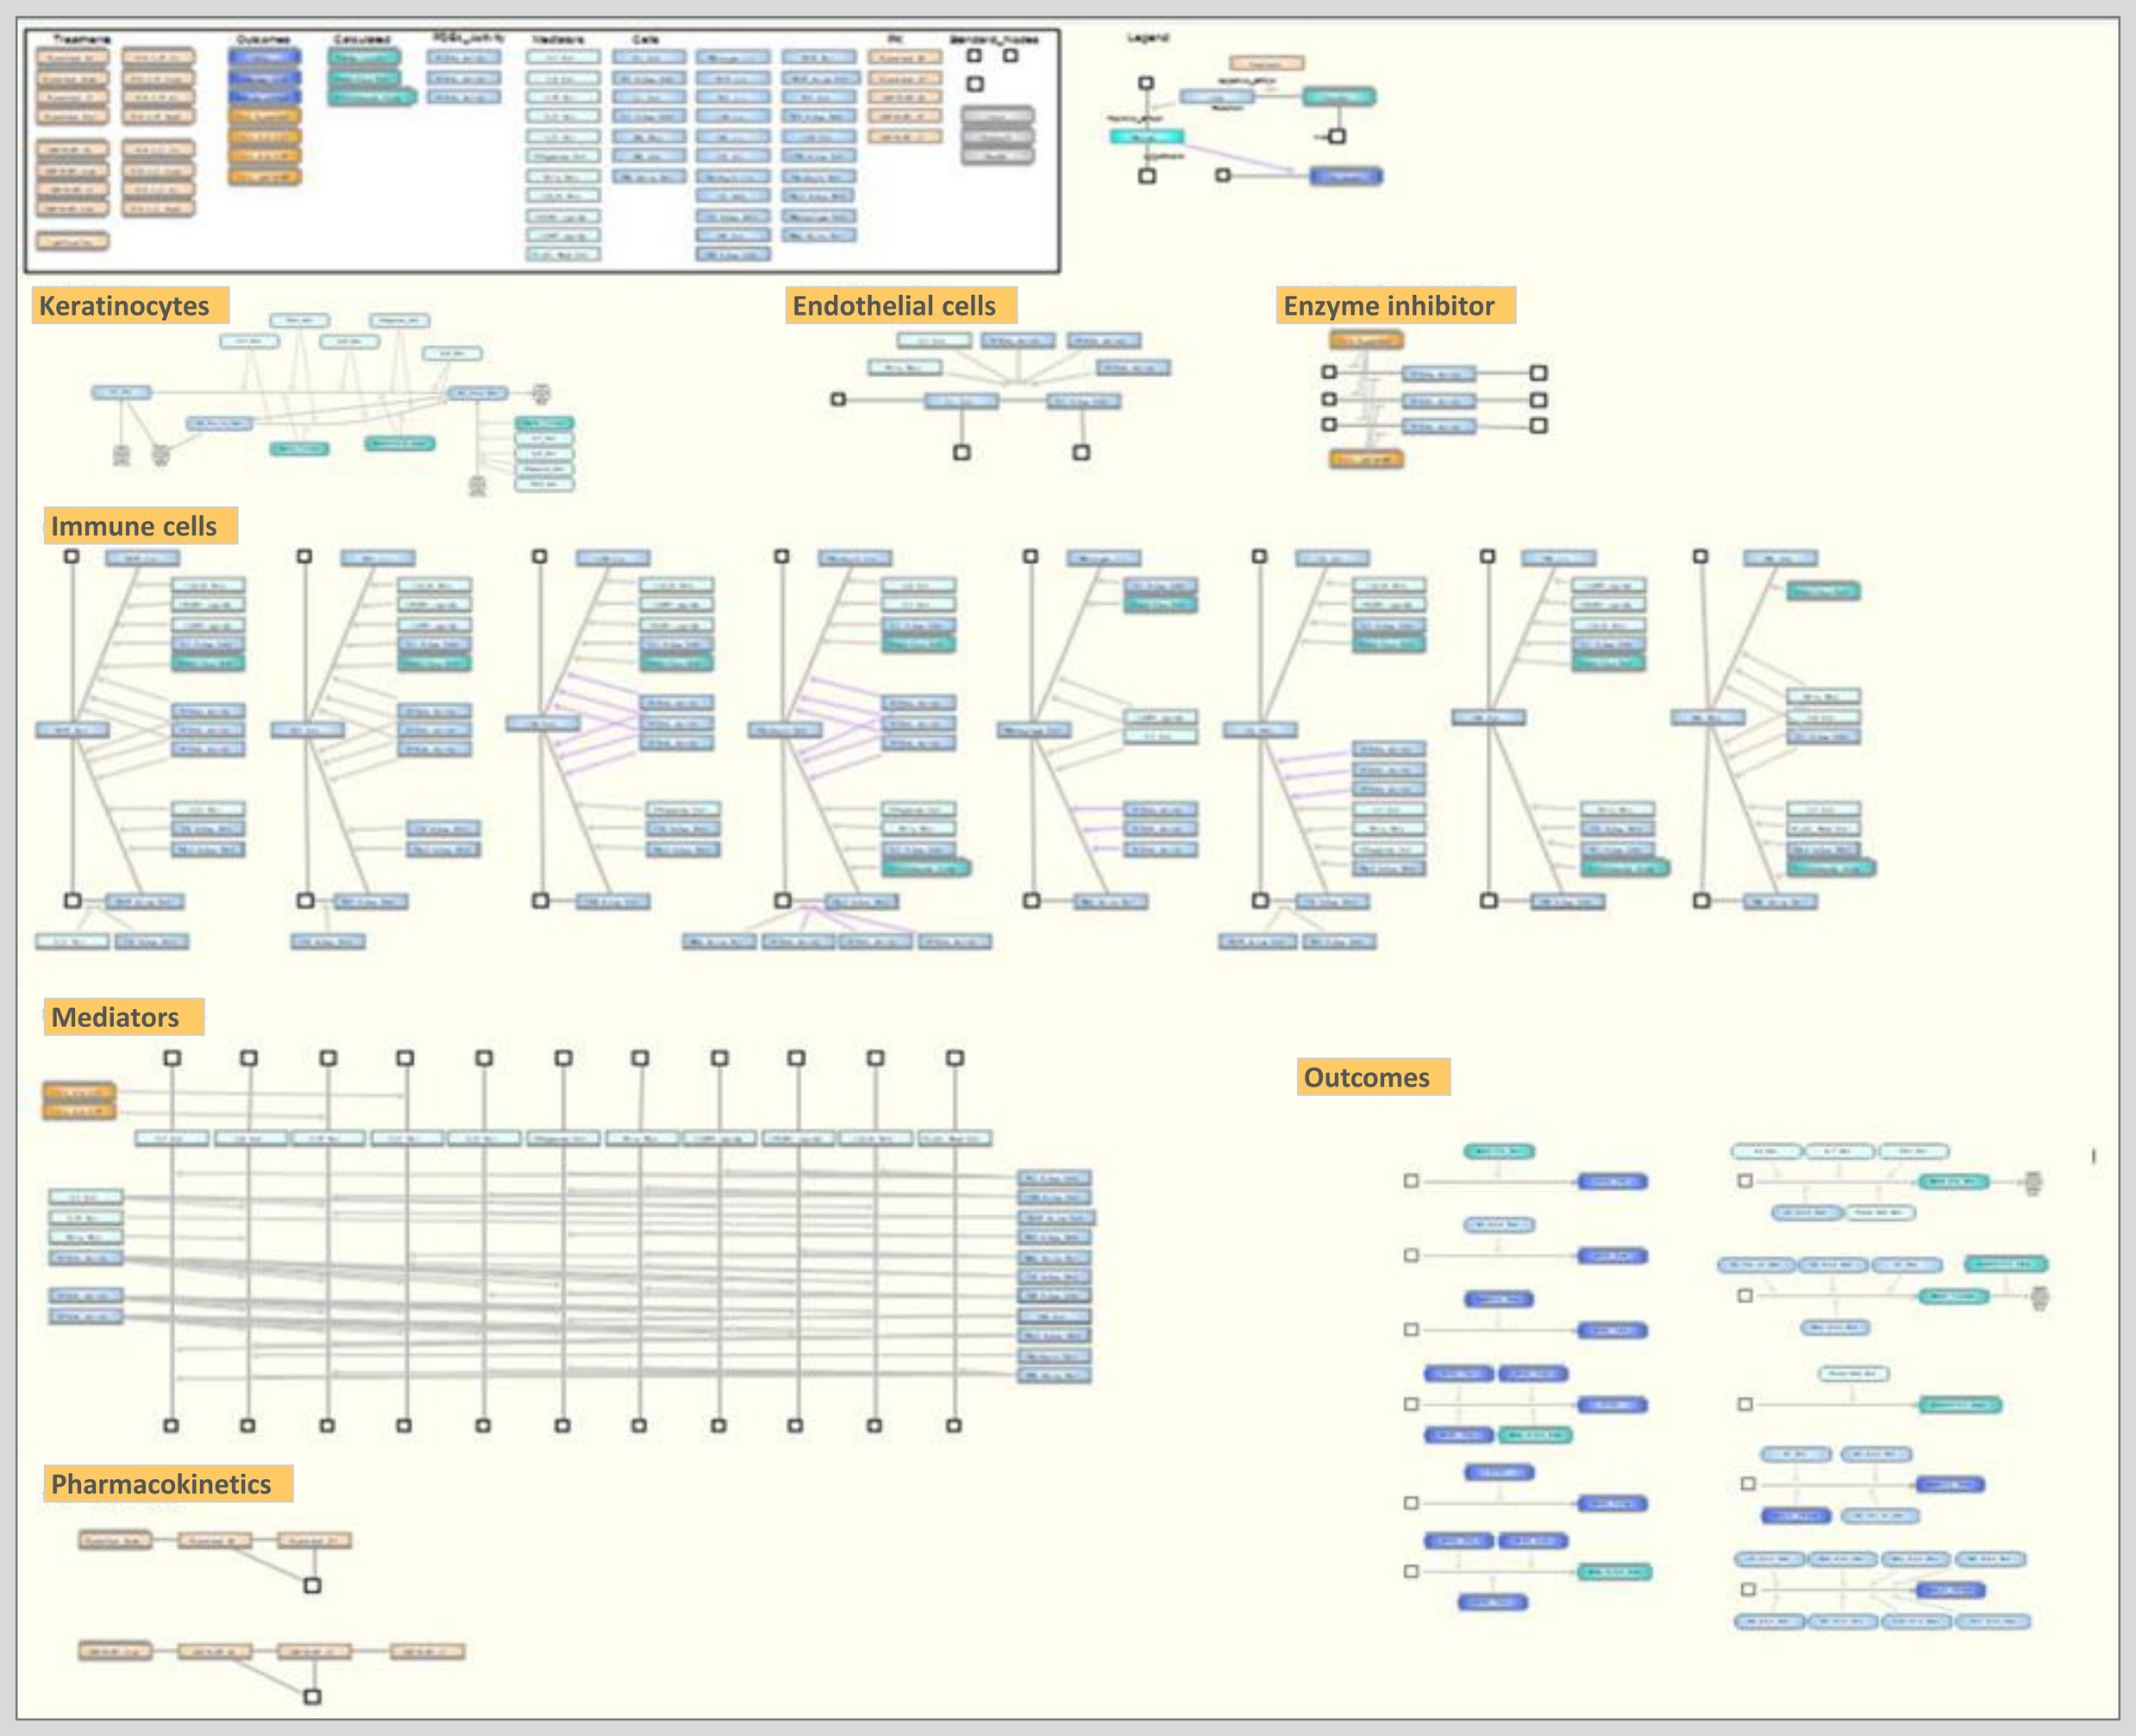
Keratinocytes
Endothelial cells
Enzyme inhibitor
Immune cells
Mediators
Outcomes
Pharmacokinetics

Figure 2. The Psoriasis PhysioPD Platform (Figure 2) represents the pathophysiology of a **single moderate/severe chronic psoriatic plaque,** including keratinocyte lifecycle and activation, recruitment and activation of immune cells, production and downstream effects of a key mediators involved in the pathophysiology of the disease, and response to treatments

The Platform includes mechanisms of action for existing treatments (e.g., anti-TNFα, anti-IL-17, and anti-IL-23, and pan-specific enzyme inhibitor), pharmacokinetics and dosing of the novel compound and the competitor drugs, and clinical outcomes related to the SPASI score such as redness, scaliness, and epidermal thickness

The Platform was developed using System Biology Workbench JDesigner®

The Platform was qualified to meet expected behaviors based on available public and proprietary data.

- Platform was qualified to match behaviors reported in the literature (e.g., epidermal thickness, immune infiltrate, SPASI score etc.) under placebo and standard of care (SOC) treatments

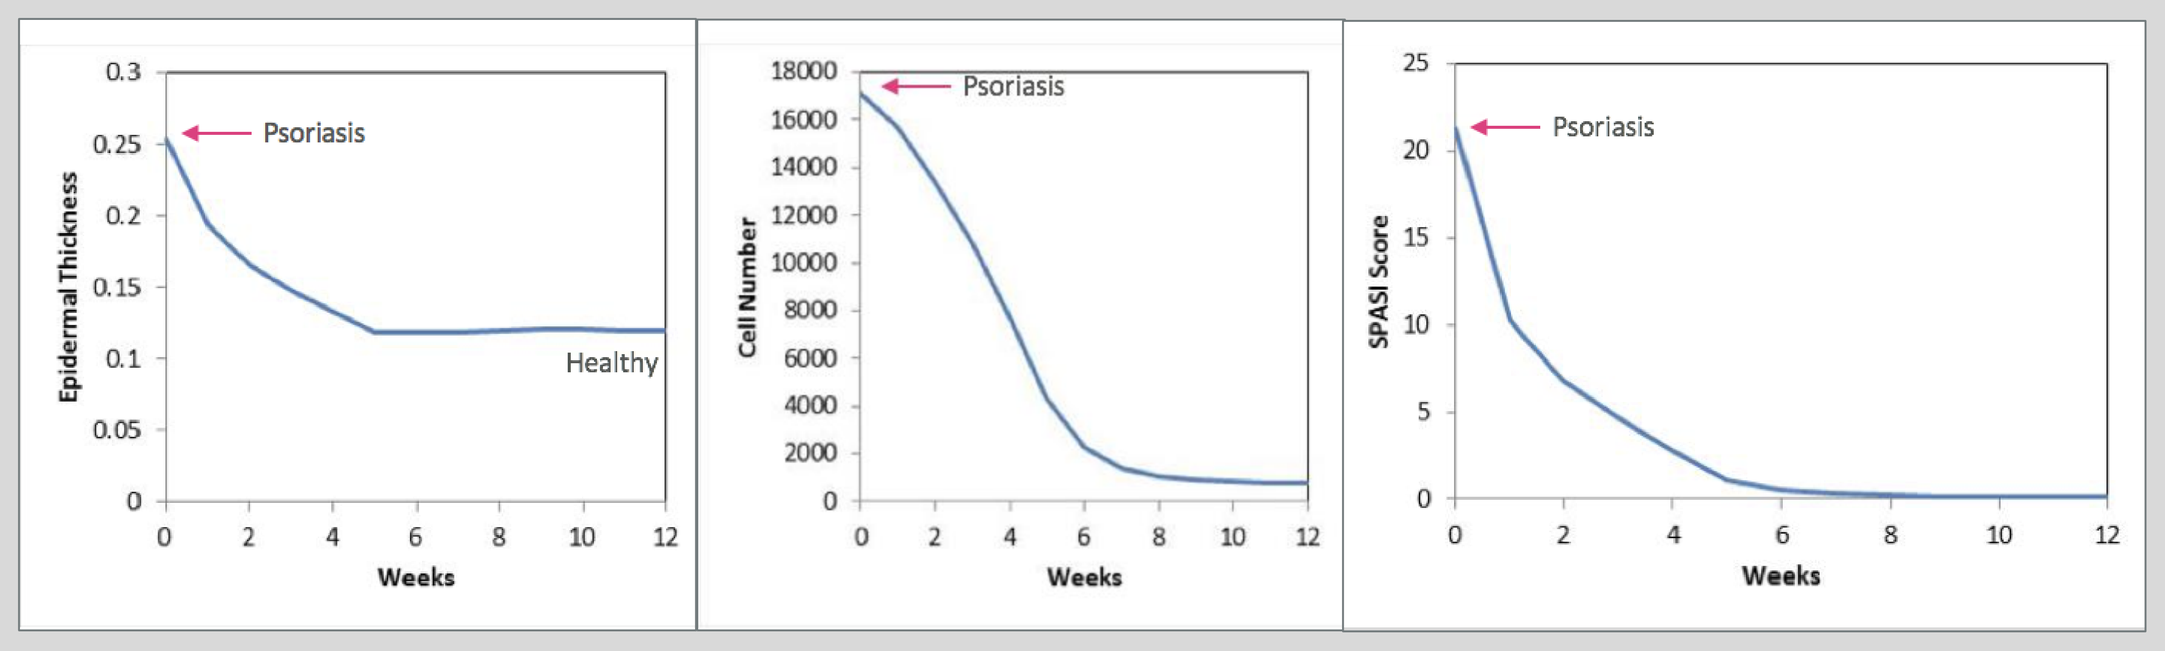
Epidermal Thickness
Psoriasis
Healthy
Weeks
Cell Number
Psoriasis
Weeks
SPASI Score
Psoriasis
Weeks

Figure 3. Example of Platform qualification: epidermal thickness (mm), inflammatory infiltrate (e.g., T cells, neutrophils, macrophages) and SPASI score decrease with simulated anti-IL-23 therapy in a psoriatic Virtual Patient. Simulated results are consistent with reported data. Over 50% decrease in epidermal thickness with anti-IL-23 treatment has been reported [4,5]. Immune infiltrate is also strongly inhibited [4,5]. Reduction is SPASI score has been shown to be >75% [4,6].

Sensitivity Analysis highlighted the key pathways most critical in the disease pathophysiology.

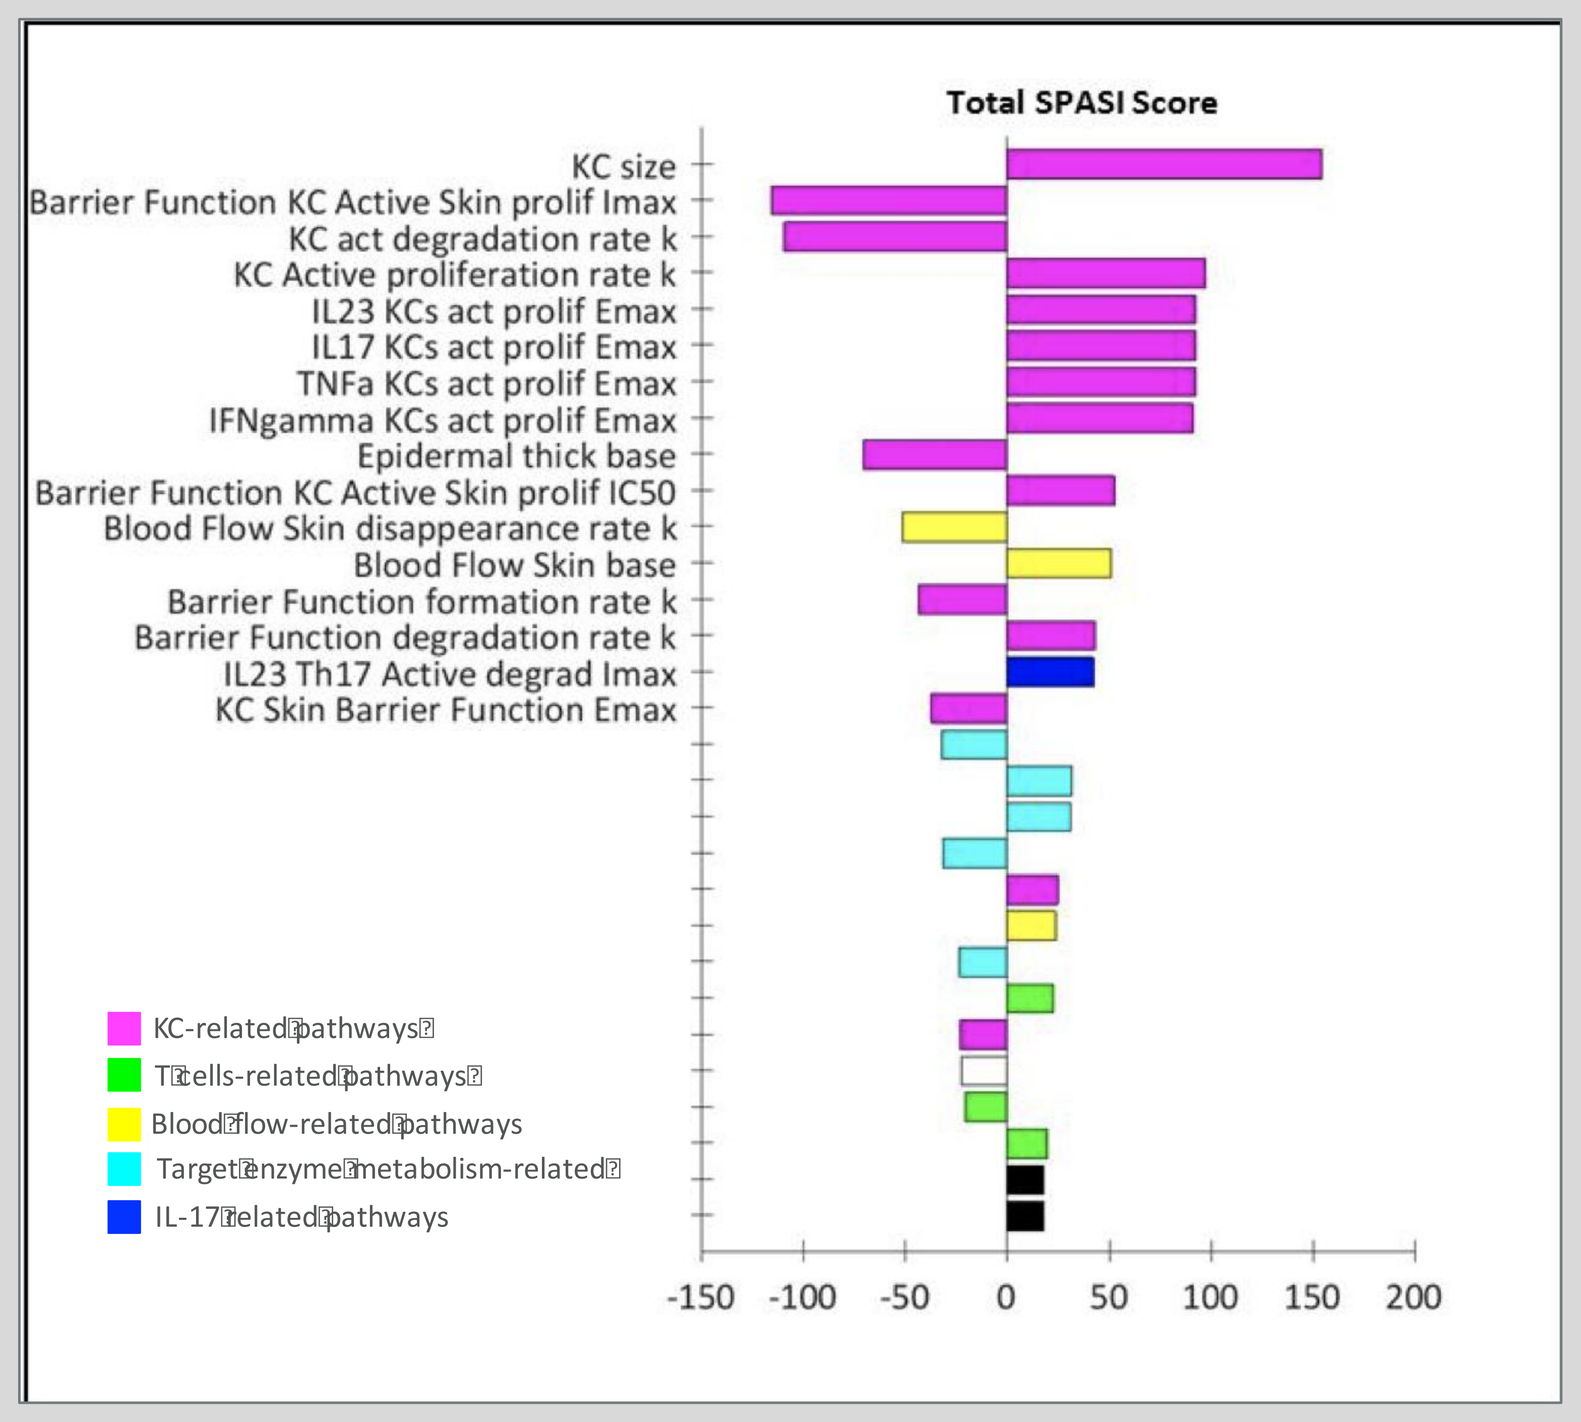
Total SPASI Score
KC size
Barrier Function KC Active Skin prolif Imax
KC act degradation rate k
KC Active proliferation rate k
IL23 KCs act prolif Emax
IL17 KCs act prolif Emax
TNFa KCs act prolif Emax
IFNgamma KCs act prolif Emax
Epidermal thick base
Barrier Function KC Active Skin prolif IC50
Blood Flow Skin disappearance rate k
Blood Flow Skin base
Barrier Function formation rate k
Barrier Function degradation rate k
IL23 Th17 Active degrad Imax
KC Skin Barrier Function Emax
KC-related pathways
T cells-related pathways
Blood flow-related pathways
Target enzyme metabolism-related
IL-17 related pathways
-150 -100 -50 0 50 100 150 200

Figure 3. Biological pathways most highly affecting the SPASI score are related to KCs, T cells, blood flow, IL-17, target enzyme effects.
Tornado diagram shows ranking the effects of parameter perturbations on biological outcomes (e.g. SPASI score shown). Other treatment-specific outcomes were evaluated. Color coding was used to assign parameters to specific metabolic pathways or cell processes.

Results

***In silico* research in Virtual Patients identified conditions under which a novel compound would be superior to the SOC treatments.**

- VPs, each representing alternative parametrizations of the model, were created to evaluate the effect of pathway uncertainties on novel compound efficacy
- Extremes of response were created by biasing all uncertainties in favor of or against the efficacy of the experimental therapy in two VPs (Figure 4)

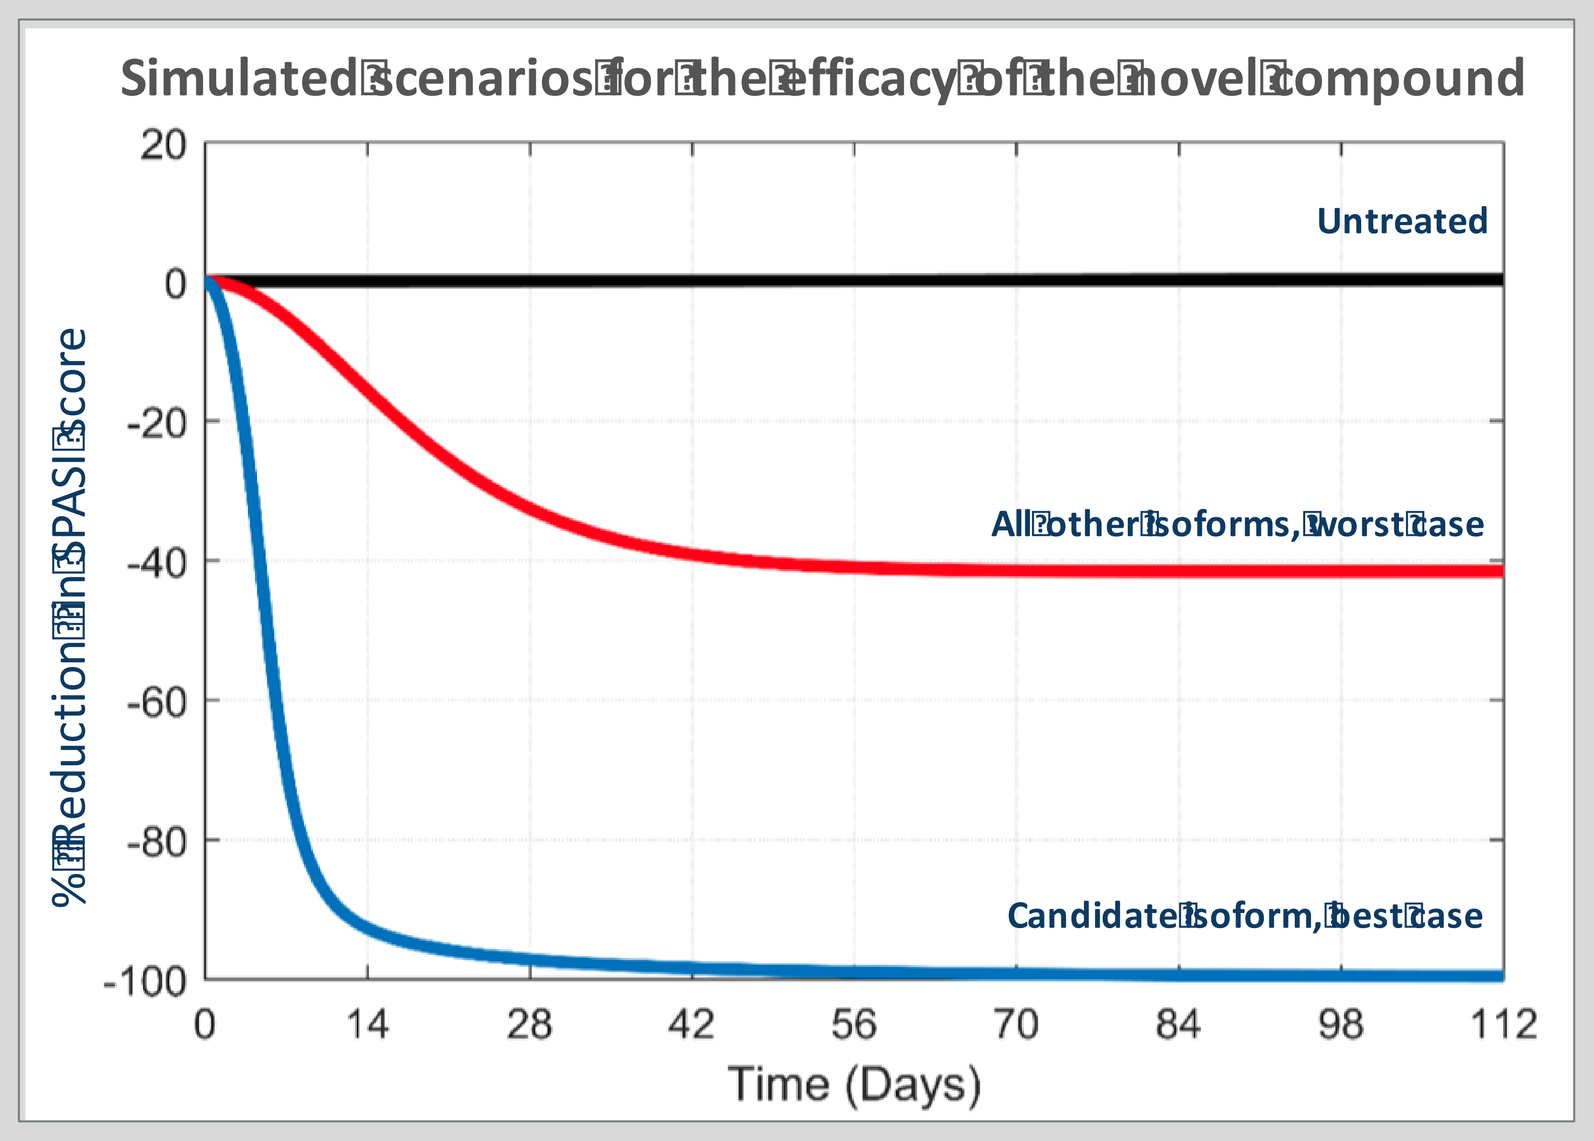
Simulated scenarios for the efficacy of the novel compound
Untreated
All other isoforms, worst case
Candidate isoform, best case
% Reduction in SPASI score
Time (Days)

Figure 4. Simulated change in SPASI score in two VPs illustrates the best and worst case scenarios for the efficacy of the novel treatment. For the best case scenario, all pathways in the Platform with uncertainties related to the candidate enzyme isoform were completely inhibited by the novel treatment. Under the worst case scenario, regulation was attributed to other enzyme isoforms.

- The key uncertainties with biggest impact on clinical outcomes were determined by SA, and included TNFα activity, and Th17, Th1 cellular effects
- Additional VPs were used to explore the impact of specific key uncertainties on the new compound efficacy (Figure 5)
- Changes in clinical outcomes under experimental therapy in VPs with different underlying assumptions illustrate the range of potential response on predicted efficacy of the novel compound
- This type of analysis identified critical uncertainties that can be resolved by additional laboratory research, reducing drug development risk

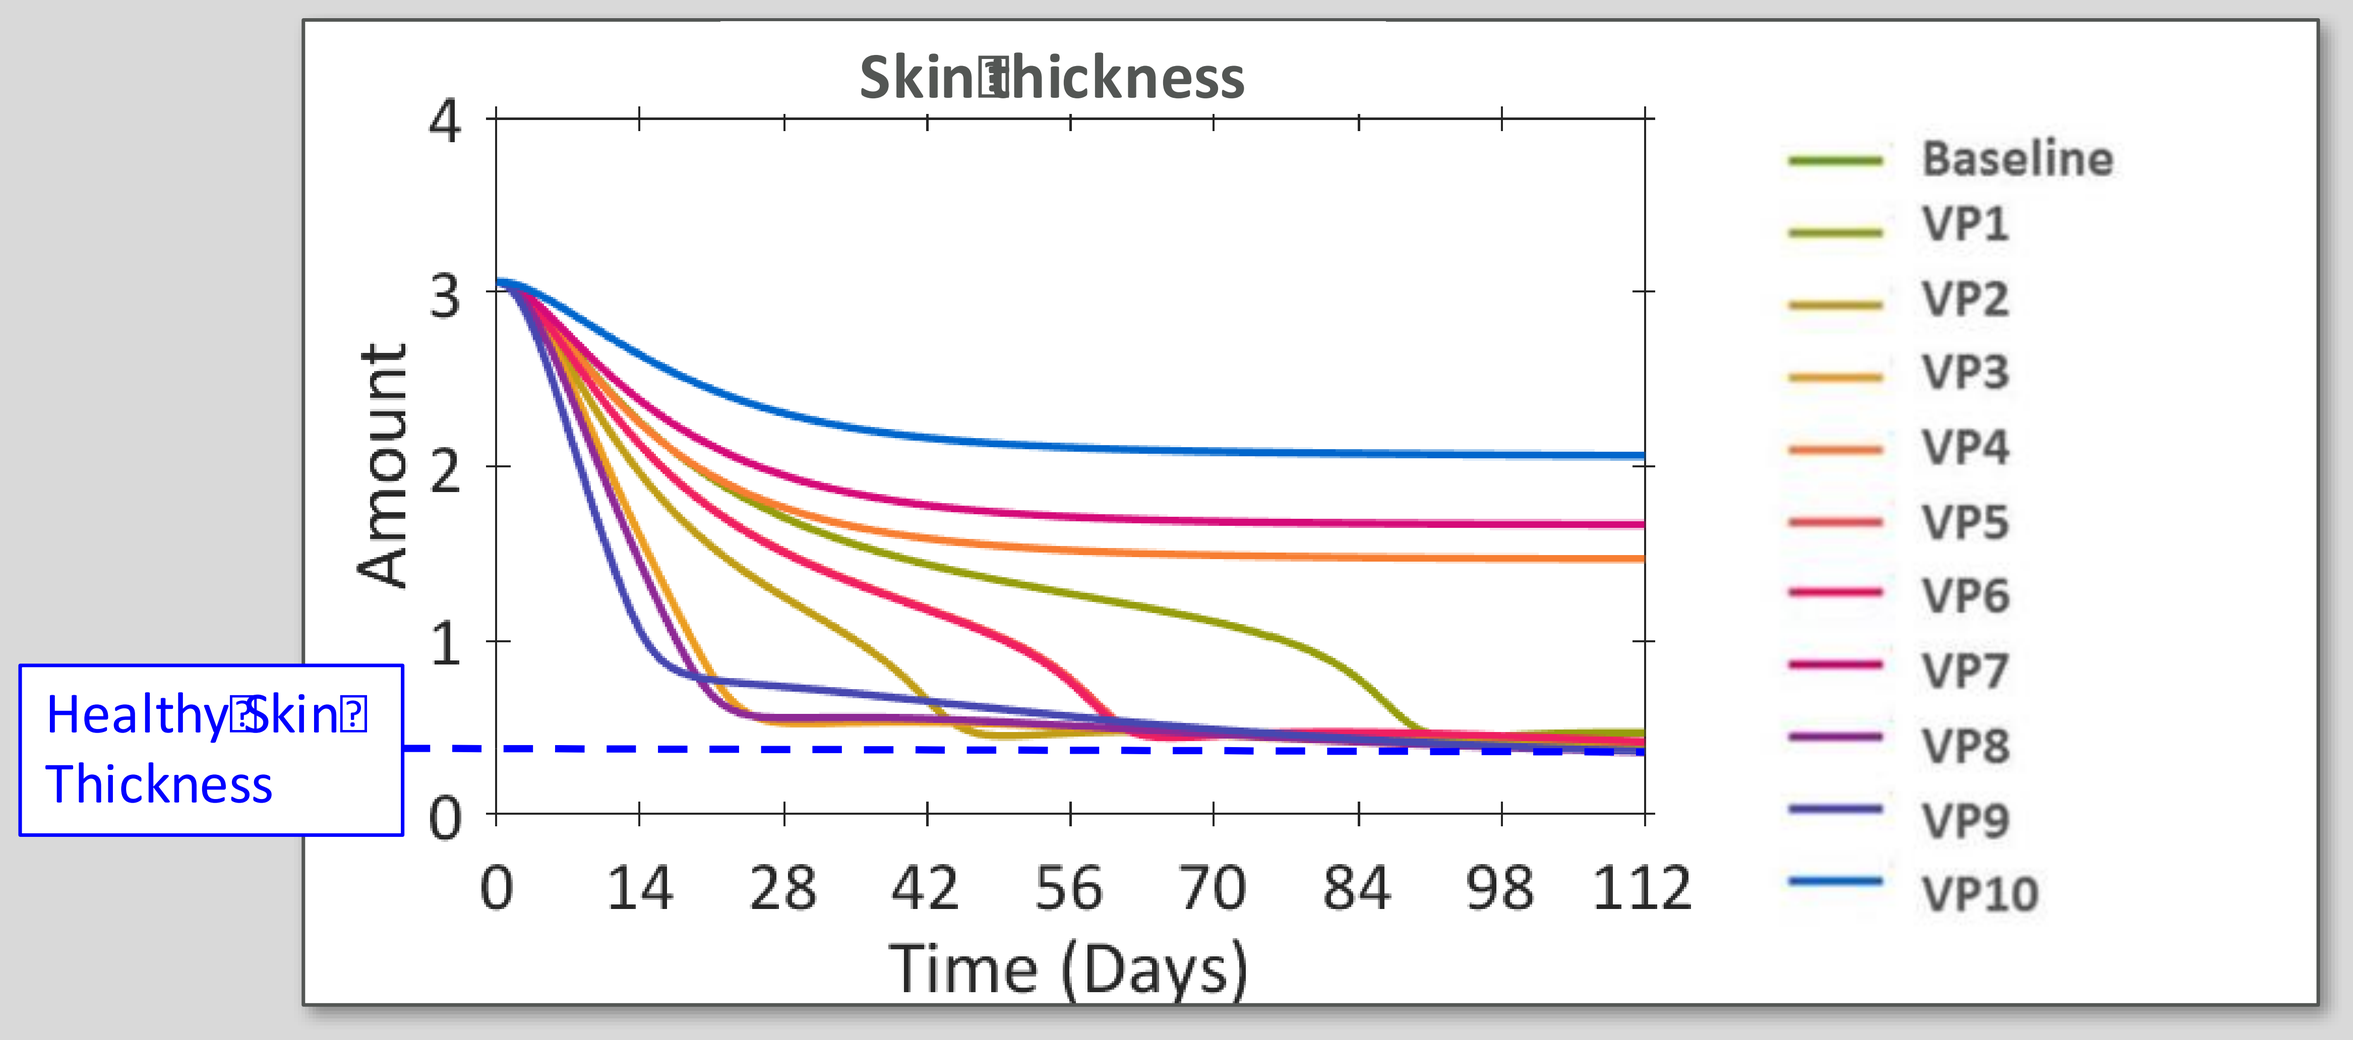
Skin thickness
Amount
Time (Days)
Healthy Skin Thickness
Baseline
VP1
VP2
VP3
VP4
VP5
VP6
VP7
VP8
VP9
VP10

Figure 5. Simulated SPASI scores reflect differential responses of ten VPs, treated with simulated experimental therapy

Conclusions

- The Psoriasis PhysioPD Platform was designed to represent the pathophysiology of a single chronic psoriatic plaque
- Platform qualification included the reported response to a variety of marketed therapies
- Sensitivity analysis highlighted the potential key drivers of disease and plaque healing
- Virtual Patients were created to evaluate the impact of target-related uncertainties
- Research in the PhysioPD Platform identified:
 - Key uncertainties related to target expression and function in immune and skin cells
 - Conditions under which novel experimental drug would be superior to the standard of care

www.rosaandco.com

For more information about this work, please contact:
Katherine Kudrycki
Rosa & Co., LLC
kkudrycki@rosaandco.com